Exploring the Clinical Course of Hair Thinning Associated With Teriflunomide: An Update to the Teriflunomide Real-World Case Series

P1113

Lori Hendin Travis, MD,[1] Annette Okai, MD,[2] Scott Jackson, MD,[3] Lisa Farnett, PharmD,[4] Steve Cavalier, MD,[4] Darren Stam, PharmD,[4] Ken Liu, PharmD,[4] Keith R Edwards, MD[5]

[1]Phoenix Neurological Associates Ltd, Phoenix, AZ, USA; [2]Multiple Sclerosis Treatment Center of Dallas, Dallas, TX, USA; [3]Baton Rouge Clinic, Baton Rouge, LA, USA; [4]Genzyme, a Sanofi company, Cambridge, MA, USA; [5]Multiple Sclerosis Center of Northeastern New York, Latham, NY, USA

OBJECTIVE

- **To illustrate the clinical course of hair thinning in the small group of patients who experience this adverse event during treatment with teriflunomide**

INTRODUCTION

- Teriflunomide is a once-daily oral immunomodulator approved for the treatment of relapsing-remitting MS
- Phase 2 and 3 placebo-controlled studies in patients with relapsing MS[1–3] and patients who experienced a first clinical episode consistent with MS[4] have demonstrated the consistent efficacy of teriflunomide on relapses,[1–4] disability progression,[1–4] and magnetic resonance imaging parameters[1,3,4]
- Teriflunomide has a well-characterized long-term safety and tolerability profile, with patients having been treated for up to 12 years in long-term extension studies[5,6]
- In a pooled analysis of the placebo-controlled trials, hair thinning occurred in 14% of patients receiving teriflunomide 14 mg vs 5% of those receiving placebo[6]
 - Most hair thinning events occurred within the first 6 months of treatment, were considered mild or moderate, and resolved without corrective treatment while patients remained on teriflunomide[6]
 - Few patients (6%) who experienced hair thinning discontinued teriflunomide treatment because of hair thinning[6]
- The assessment and characterization of teriflunomide-associated hair thinning in real-world experiences could help set appropriate expectations for patients before initiating treatment with teriflunomide

METHODS

Participants

- Patients with relapsing-remitting MS who reported hair thinning to healthcare professionals (HCPs) during treatment with teriflunomide were eligible for inclusion

Study Design

- This was an observational real-world project conducted at 9 MS centers in the United States, with a data cutoff of May 14, 2015
- During 2 office visits, 1 at the onset of hair thinning and another at follow-up, HCPs and patients completed hair thinning questionnaires that ranked severity as mild/moderate/severe or from 0 to 10, respectively
- Patients were photographed in 5 standard views (anterior, posterior, left lateral, right lateral, and anterior superior) and an optional manipulated view with their hair pulled back
- A post hoc analysis of the hair photos was performed by a dermatologist to assess the severity of hair loss using a scale from 1 (least severe hair thinning) to 10 (most severe hair thinning)[7]

RESULTS

Baseline Characteristics

- At data cutoff, 38 patients had completed follow-up visits
- Baseline characteristics are shown in **Table 1**

Table 1. Baseline Patient Characteristics[a]

	Patients (N=38)
Female	37 (97)
Age, mean (SD), years	52 (12)
Caucasian/white	32 (84)
No history of hair thinning	33 (87)
Use of concomitant medications associated with hair thinning	26 (68)
EDSS, mean (SD) EDSS, median (min:max)	2.60 (1.27) 2.5 (0.0:5.5)

[a]Data presented as n (%) unless otherwise stated.
SD, standard deviation.

Hair Thinning

- **Figure 1** presents examples of hair thinning reported in this study
- The mean time from starting teriflunomide treatment to onset of hair thinning was 77 days (<3 months), and from onset to follow-up visits was 270 days
- At onset, mean score for patient perception of severity was 5.1/10 (n=38), whereas mean dermatologist-rated severity was 3/10 (n=31). HCPs classified most cases as mild (63%) or moderate (34%), with a single severe case in a 33-year-old female with a prior history of drug-induced hair loss (**Table 2**)
- Complete/near-complete resolution or marked improvement was reported by 79% of patients at follow-up
 - Only 1 patient described minimal improvement (hair thinning rated as mild by HCP, 1/10 by dermatologist, and 2/10 by patient at both visits)
- HCPs reported less severity in follow-up visits, and the dermatologist assessment of severity remained low, 1.5/10[7]
- Teriflunomide treatment was discontinued in 5 patients:
 - Three permanently, 1 each due to hair thinning, rash, and gastrointestinal upset
 - Two temporarily, 1 due to hair thinning and 1 due to gastrointestinal upset

CONCLUSIONS

- **Consistent with observations from the clinical trial program, reported hair thinning events in patients receiving teriflunomide were typically mild, occurred at a low incidence rate and within 3 months, and usually resolved without treatment discontinuation**
- **Patient-perceived severity at onset and follow-up visits were generally more severe than assessments by either the treating HCP or the dermatologist**
- **This small case series provides patients' own perspectives and provides real-world photographs of hair thinning associated with teriflunomide**

Figure 1. Examples of Hair Thinning at Onset and Follow-up

	Patient 1		Patient 2		Patient 3		Patient 4	
	Onset	Follow-up	Onset	Follow-up	Onset	Follow-up	Onset	Follow-up
	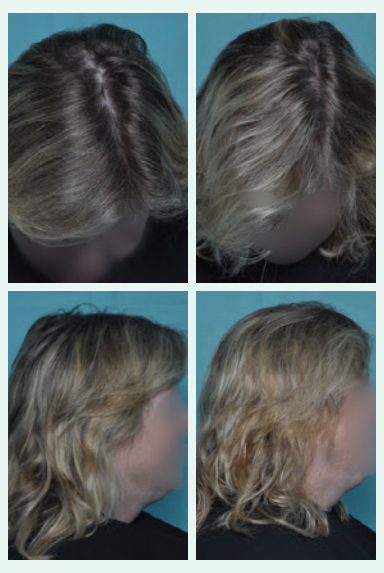		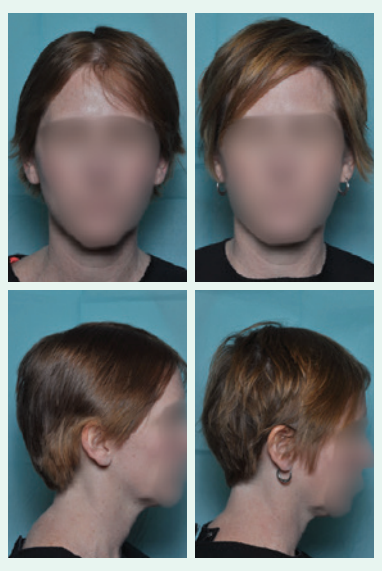		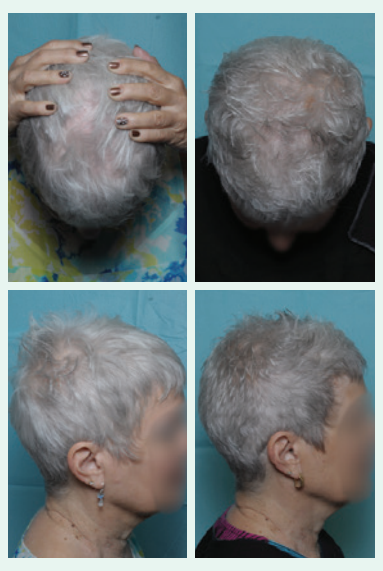		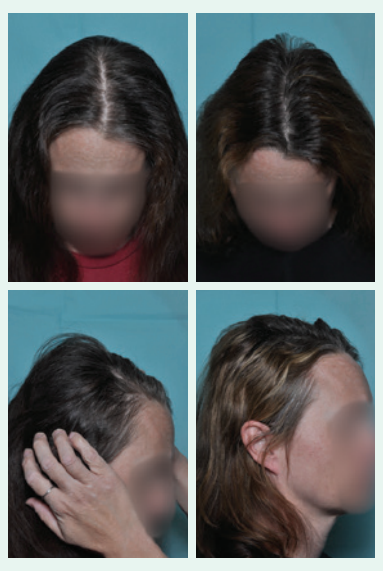	
Age, years	57		38		70		44	
Time from first dose of teriflunomide to onset of hair thinning, days	170		62		112		83	
Time from onset to follow-up visit, days	303		207		244		244	
Severity Assessment	**Onset**	**Follow-up**	**Onset**	**Follow-up**	**Onset**	**Follow-up**	**Onset**	**Follow-up**
Patient	3/10	1/10; complete/ near-complete resolution	3/10	3/10; markedly improved	5/10	3/10; markedly improved	5/10	5/10; complete/ near-complete resolution
HCP	Mild	Mild	Mild	Mild	Mild	Mild	Mild	Moderate
Dermatologist	2/10	1/10	3/10	2/10	2/10	1/10	3/10	2/10
Location of hair thinning reported by patient	Top		Temporal (left and right), frontal (left, center, and right), top, back		Top		Temporal (left and right), frontal (left, center, and right), top, back	
Teriflunomide continued? (Yes/No)	Yes		Yes		Yes		Yes	
	Patient 5		**Patient 6**		**Patient 7**		**Patient 8**	
	Onset	Follow-up	Onset	Follow-up	Onset	Follow-up	Onset	Follow-up
	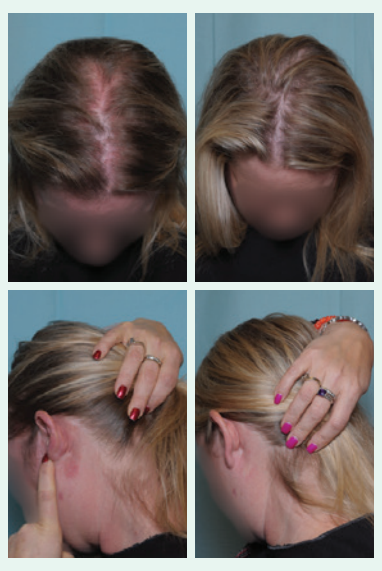		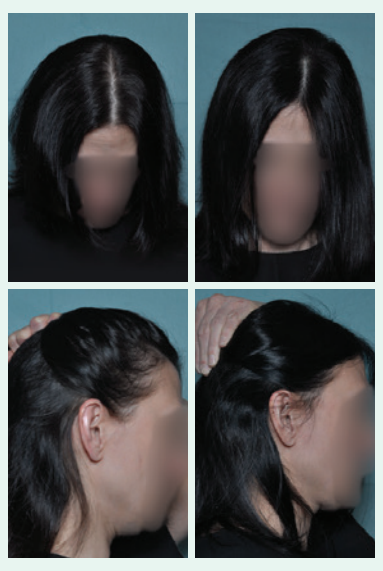		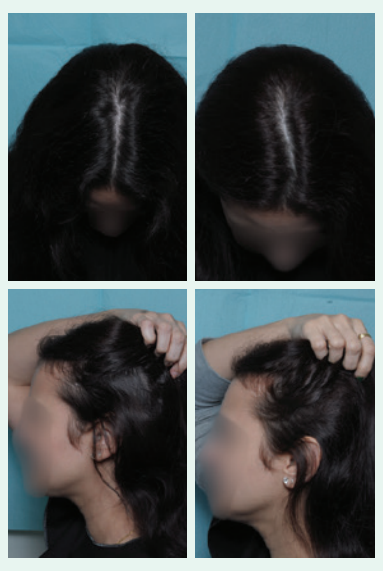		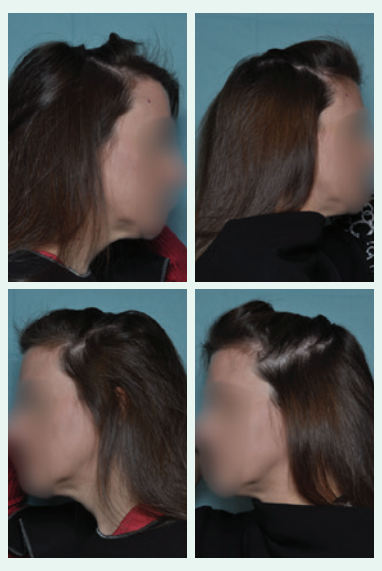	
Age, years	45		46		42		46	
Time from first dose of teriflunomide to onset of hair thinning, days	30		109		56		354	
Time from onset to follow-up visit, days	338		258		158		247	
Severity Assessment	**Onset**	**Follow-up**	**Onset**	**Follow-up**	**Onset**	**Follow-up**	**Onset**	**Follow-up**
Patient	7/10	1/10; complete/ near-complete resolution	7/10	2/10; markedly improved	7/10	4/10; somewhat improved	7/10	8/10; complete/ near-complete resolution
HCP	Moderate	Mild	Moderate	Mild	Moderate	Mild	Moderate	Moderate
Dermatologist	2/10	1/10	3/10	1/10	3/10	1/10	3/10	2/10
Location of hair thinning reported by patient	Temporal left, top, other (left side behind ear)		Not provided		Temporal (left and right)		All over	
Teriflunomide continued? (Yes/No)	No, discontinued due to rash		Yes		Yes		Yes	

HCP, healthcare professional.

Table 2. Summary of Hair Thinning AEs as Assessed by Patients, HCPs, and Dermatologist's Review

Study Site Assessment		**Onset Visit (n=38)**	**Follow-up Visit (n=38)**
Patient perception of severity, mean (range)		5.0 (2–10)	3.5 (1–9)
Patient rating of resolution, n (%)	Complete/near-complete	-	14 (37)
	Markedly improved	-	16 (42)
	Somewhat improved	-	7 (18)
	None/minimal	-	1 (3)
HCP perception of severity, n (%)	Mild	24 (63)	31 (82)
	Moderate	13 (34)	7 (18)
	Severe	1 (3)	0
Post Hoc Analysis of Hair Photos[7]		**Onset Photo (n=31)**	**Follow-up Photo (n=31)**
Dermatologist's perception of severity, mean (range)		3.0 (2–5)	1.5 (1–3)

AE, adverse event; HCP, healthcare professional.

References

1. O'Connor et al. *Neurology*. 2006;66:894.
2. O'Connor et al. *N Engl J Med*. 2011;365:1293.
3. Confavreux et al. *Lancet Neurol*. 2014;13:247.
4. Miller et al. *Lancet Neurol*. 2014;13:977.
5. Leist et al. Poster P7.268, AAN 2015.
6. Leist et al. Poster P2.203, AAN 2014.
7. Jackson et al. Poster P1162, EAN 2015.

Acknowledgments and Disclosures

This poster was reviewed by Larisa Miller, PharmD, Alex Lublin, PhD, and Richard Able, PhD of Genzyme, a Sanofi company. Editorial support for this poster was provided by Sarah Hauze, of Fishawack Communications, and was funded by Genzyme.

LHT: Consulting fees (Acorda, Biogen Idec, EMD Serono, Genzyme, Novartis, Pfizer, Mallinckrodt); research support (Biogen Idec, EMD Serono, Genzyme). **AO:** Consulting fees (Bayer, Biogen Idec, EMD Serono, Genzyme, Questcor, Teva Neuroscience); research support (Genzyme, Novartis, Sun Pharma). **SJ:** Nothing to disclose. **LF, DS,** and **KL:** Employees of Genzyme. **SC:** Employee of Genzyme, with ownership interest. **KRE:** Consulting fees (Biogen, Genzyme); speakers bureaus (Biogen, Genzyme, Novartis); research support (Biogen, Eisai, Eli Lilly, Forum Pharmaceuticals, Genentech, Genzyme, Hoffman-La Roche, Merz Pharmaceuticals, Novartis, Pfizer, Vaccinex).

Teriflunomide is approved in many countries, including the US and the European Union, for the treatment of relapsing multiple sclerosis or relapsing-remitting multiple sclerosis.
This material may contain information that is outside of the approved labeling in some countries.

Presented at the 31st Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS), October 7–10, 2015, Barcelona, Spain
Funding provided by Genzyme, a Sanofi company