Interim Analysis of A Pilot Trial of Natalizumab in Inclusion Body Myositis

Todd D Levine, MD and David S. Saperstein, MD

Phoenix Neurological Associates, Phoenix AZ 85018

Introduction

Inclusion Body Myositis (IBM) is the most common form of inflammatory myopathy in people over age 50. Yet in contrast to polymyositis or dermatomyositis, there are no effective therapies for IBM. The muscle biopsies of patient with IBM show a T cell predominant inflammatory infiltrate suggesting a novel agent that prevents T-cell emigration out of the vasculature may be beneficial in the therapy of patients with IBM. Natalizumab is a synthesized monoclonal antibody that blocks the interaction of VLA-4, found on leukocytes, and VCAM-1 found on the endothelial surface of the blood vessels. Natalizumab is approved for the treatment of patient with relapsing remitting MS and is very effective in preventing the migration of leukocytes out of the blood vessels.

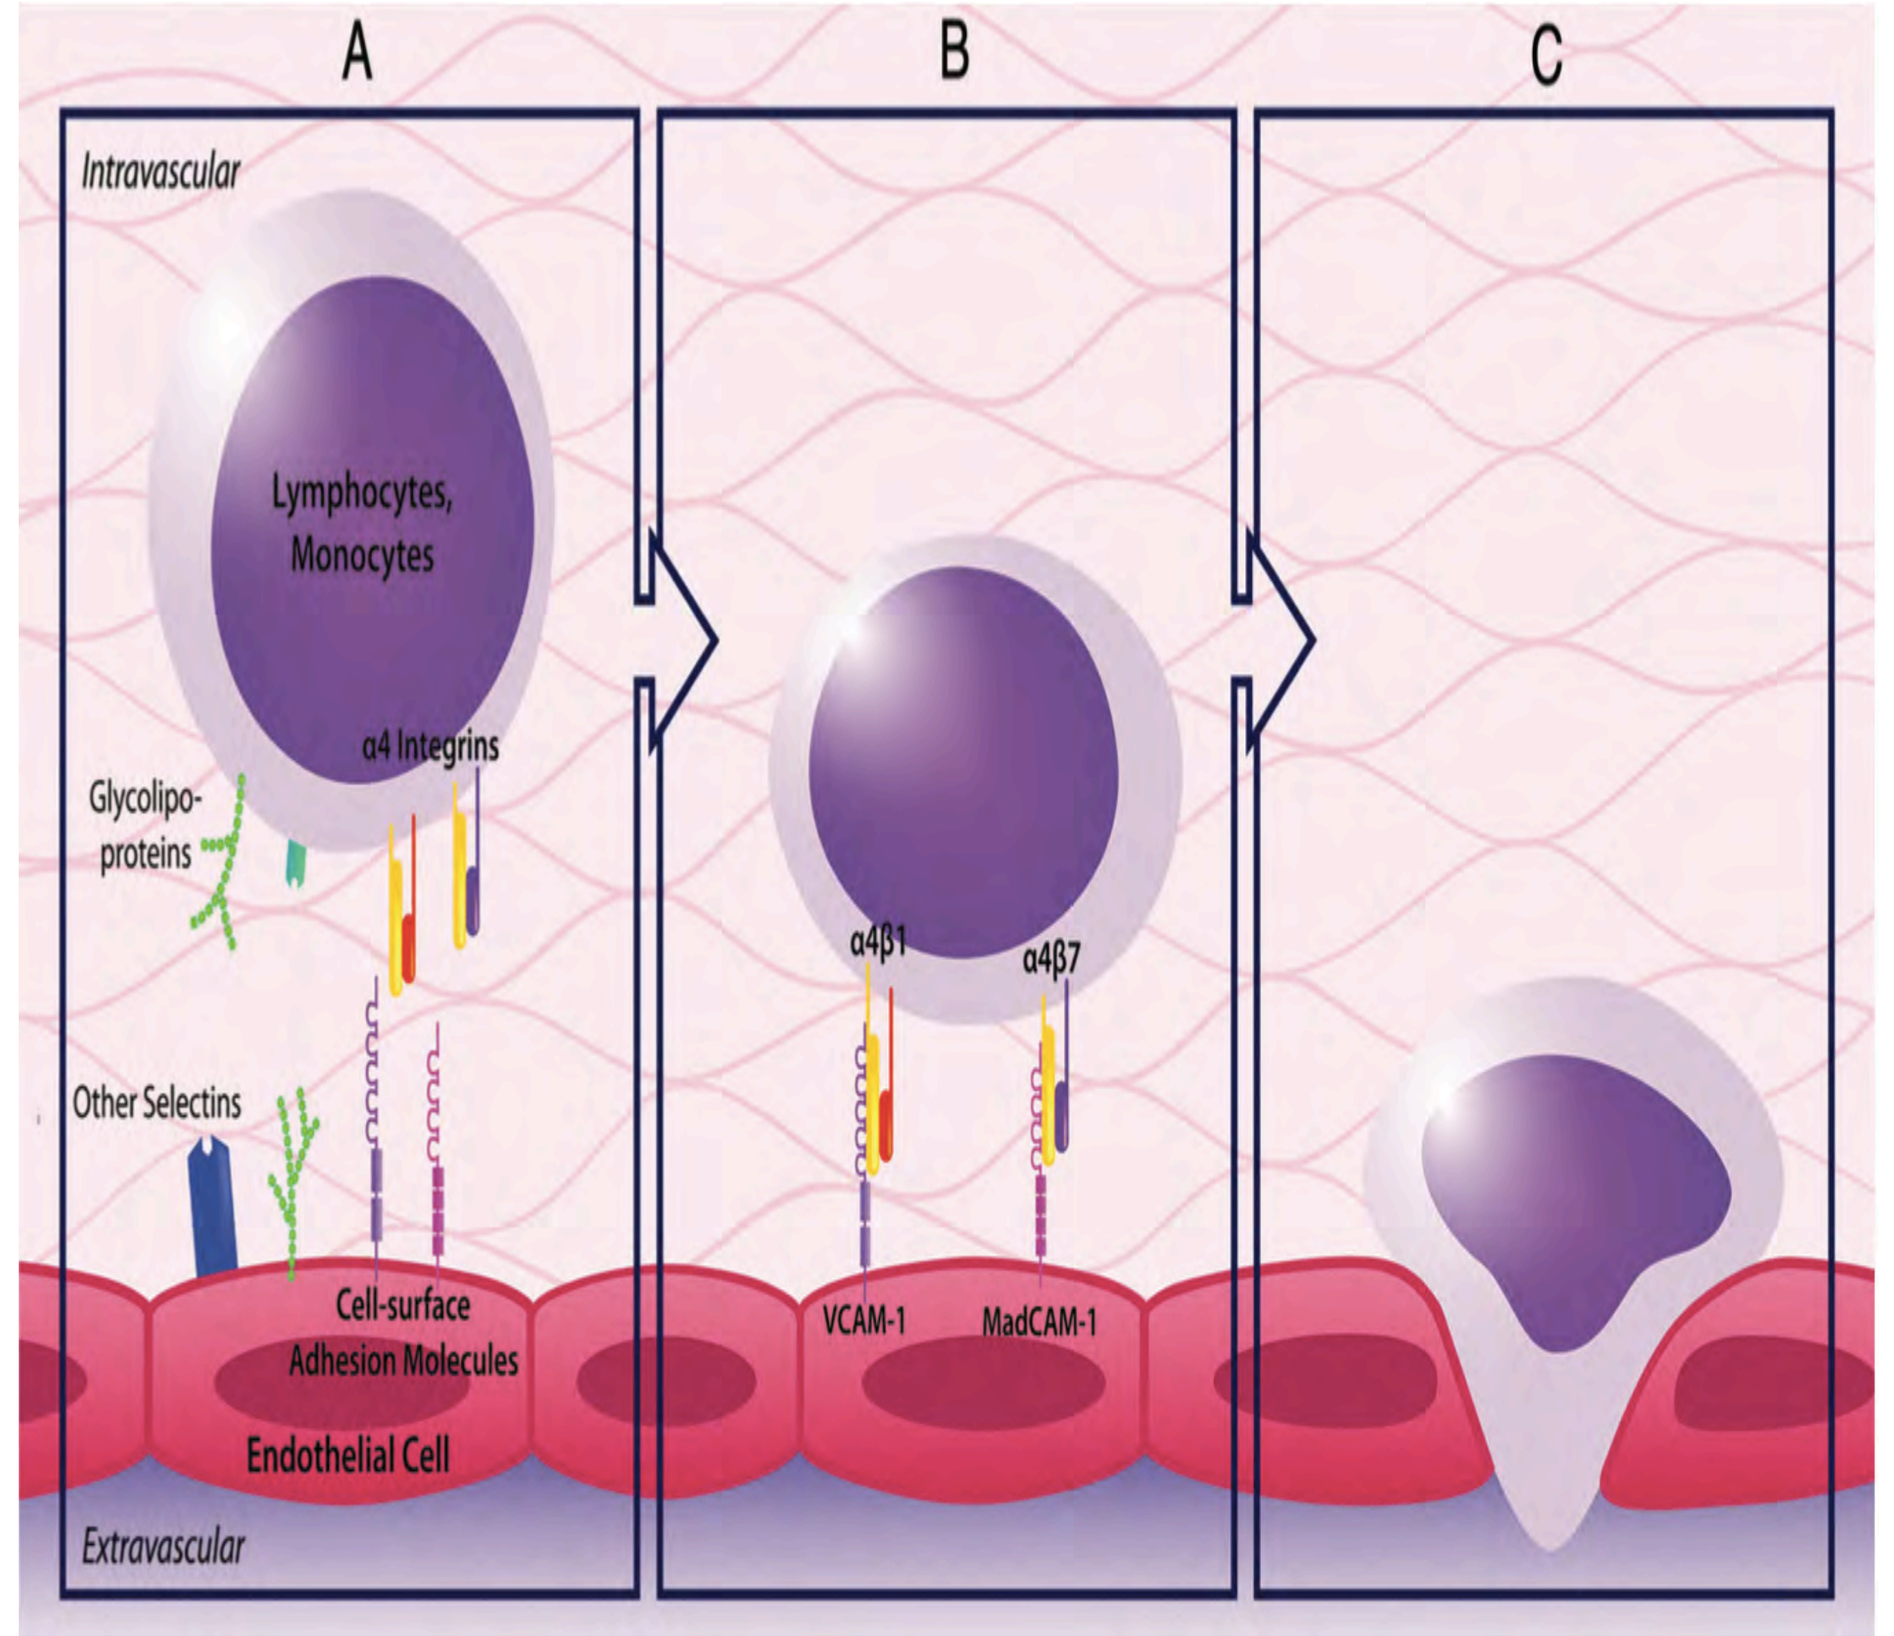

A
B
C
Intravascular
Lymphocytes, Monocytes
α4 Integrins
Glycolipo-proteins
Other Selectins
α4β1
α4β7
Cell-surface Adhesion Molecules
VCAM-1
MadCAM-1
Endothelial Cell
Extravascular

Figure 1. Mechanism of leukocyte migration through endothelium into muscle

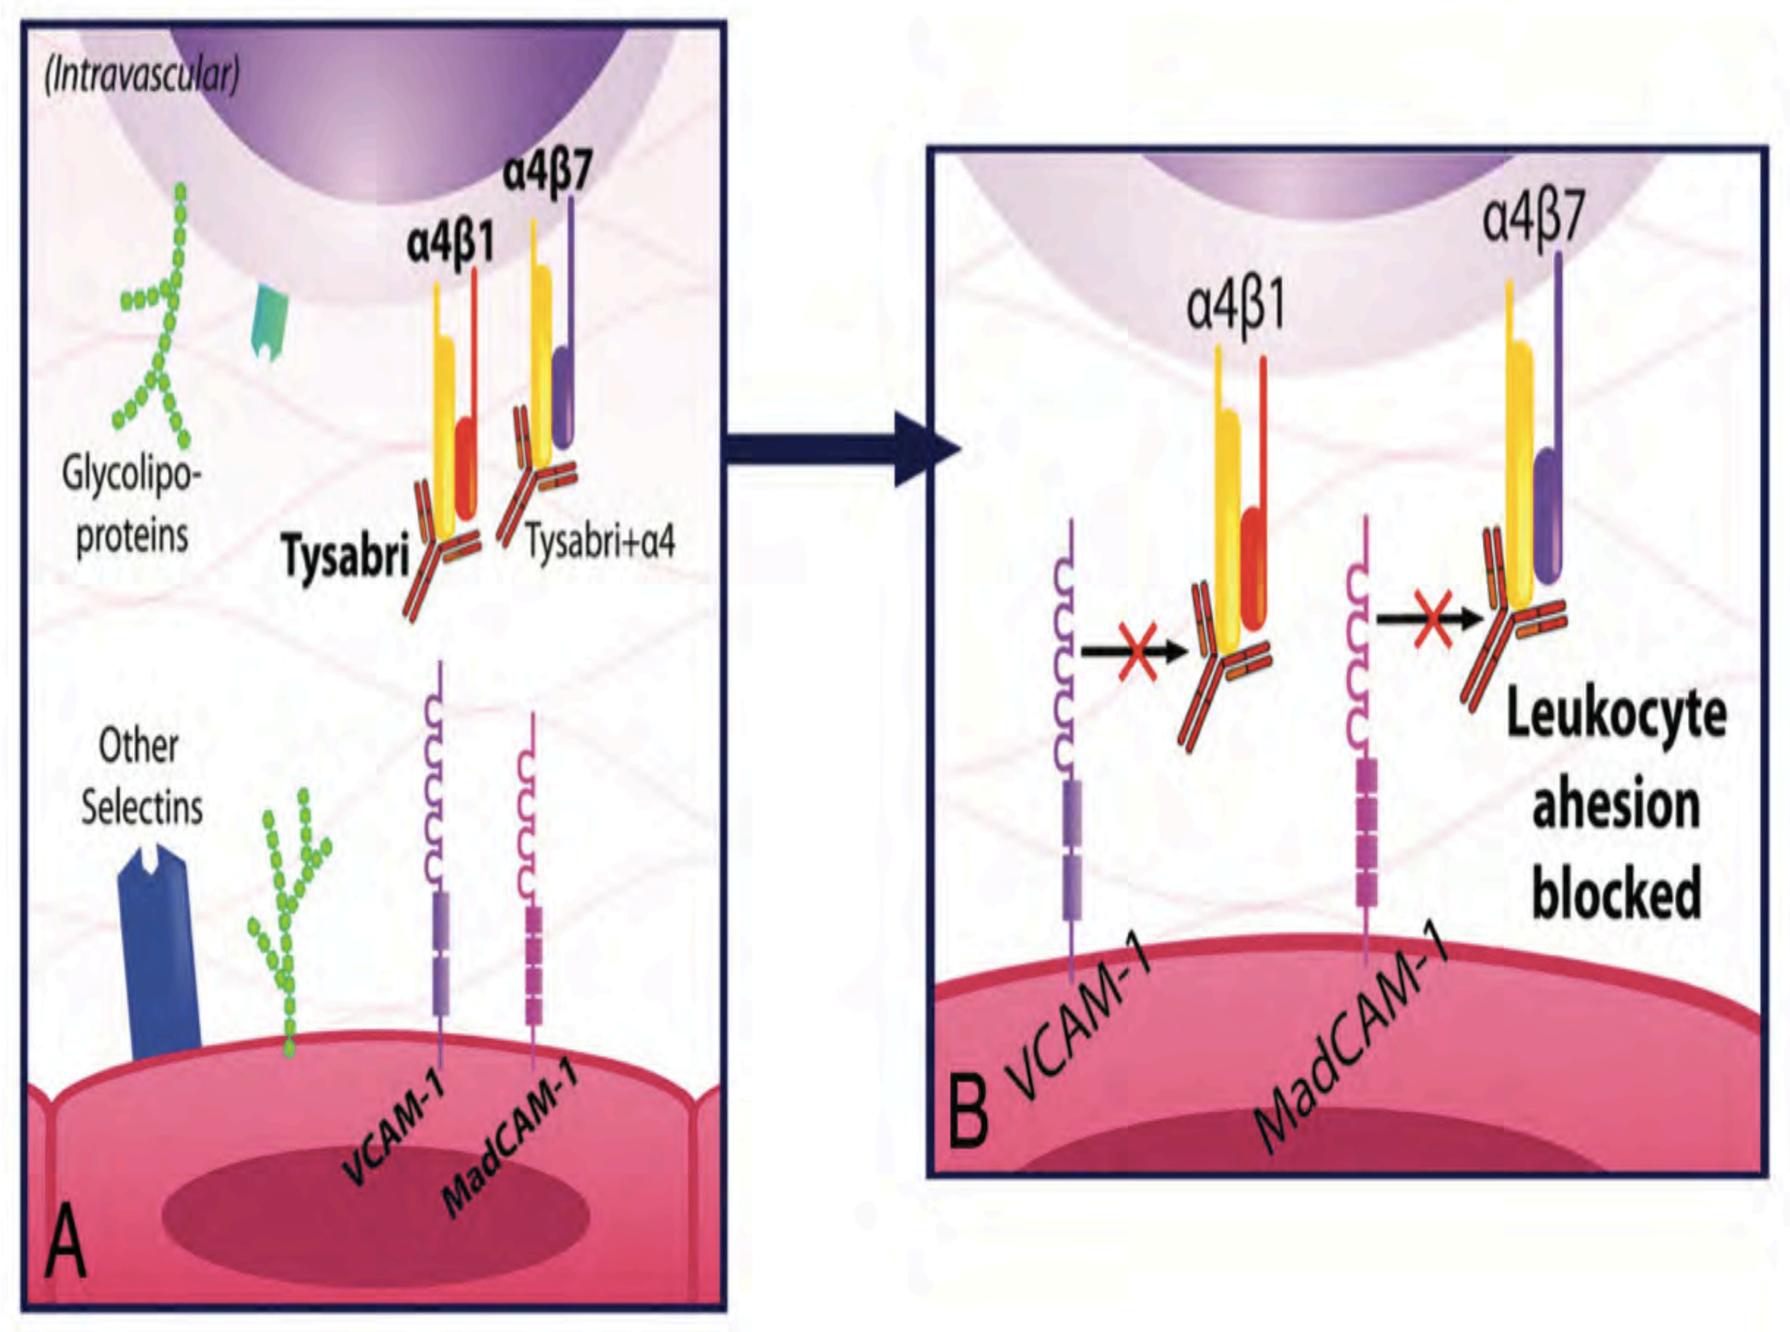

(Intravascular)
α4β7
α4β1
Glycolipo-proteins
Tysabri
Tysabri+α4
Other Selectins
VCAM-1
MadCAM-1
A
α4β1
α4β7
Leukocyte ahesion blocked
B VCAM-1
MadCAM-1

Figure 2. Natalizumab binds to VLA-4 on the surface of leukocytes which blocks binding To VCAM-1

Objectives

We proposed a phase I trial to look at the safety of Natalizumab in patients with IBM. Our hypothesis was that blocking the interaction of VLA-4 with VCAM-1 would result in the inability of lymphocyte translocation across the endothelium. This would abolish T-cell infiltration in muscles and improve the course of the disease for patients with IBM.

Methods

2 patients aged 74 and 78, both male, had biopsy proven IBM and significant muscle weakness. They were screened and enrolled in the study. Patients signed an Informed Consent and the study was reviewed by the Western IRB. All patients had a baseline quadriceps muscle biopsy performed within 4 weeks of the first dose of Natalizumab. Patients received 300 mg of Natalizumab intravenously every month for 6 months. Monitoring of CBC, CMP, CPK was performed at baseline and at months 3 and 6. Patients had quantitative muscle testing using hand held dynomometry, Manual Muscle Testing, and routine neurologic and physical exams at baseline and at 3 and 6 months. Quality of life measures were also assessed at baseline and at 3 and 6 months. Patients had a repeat biopsy after six months of natalizumab. All adverse events were recorded.

Results

To date two of the six proposed patients have been enrolled and have completed six months of therapy. There were no reported adverse events or SAEs. At three months both patients demonstrated improved strength on the Manual Muscle Testing. MMT 26 improved by 13.7% and 11.8% respectively. Patient 1 reported subjectively feeling stronger with fewer falls and handheld dynomometry showed a 24% improvement in knee extension. Patient 2 did not note any significant change in his function or QOL at 3 months. At six months both patients MMT was the same as baseline and there was no improvement or decline in quantitative dynomometry. There was also no meaningful change QOL. CPK at baseline was minimally elevated in both patients (283, 342) and was unchanged after 6 months of Natlazumab.
The muscle biopsies performed at baseline and 6 months showed a marked reduction in endomysial inflammation.

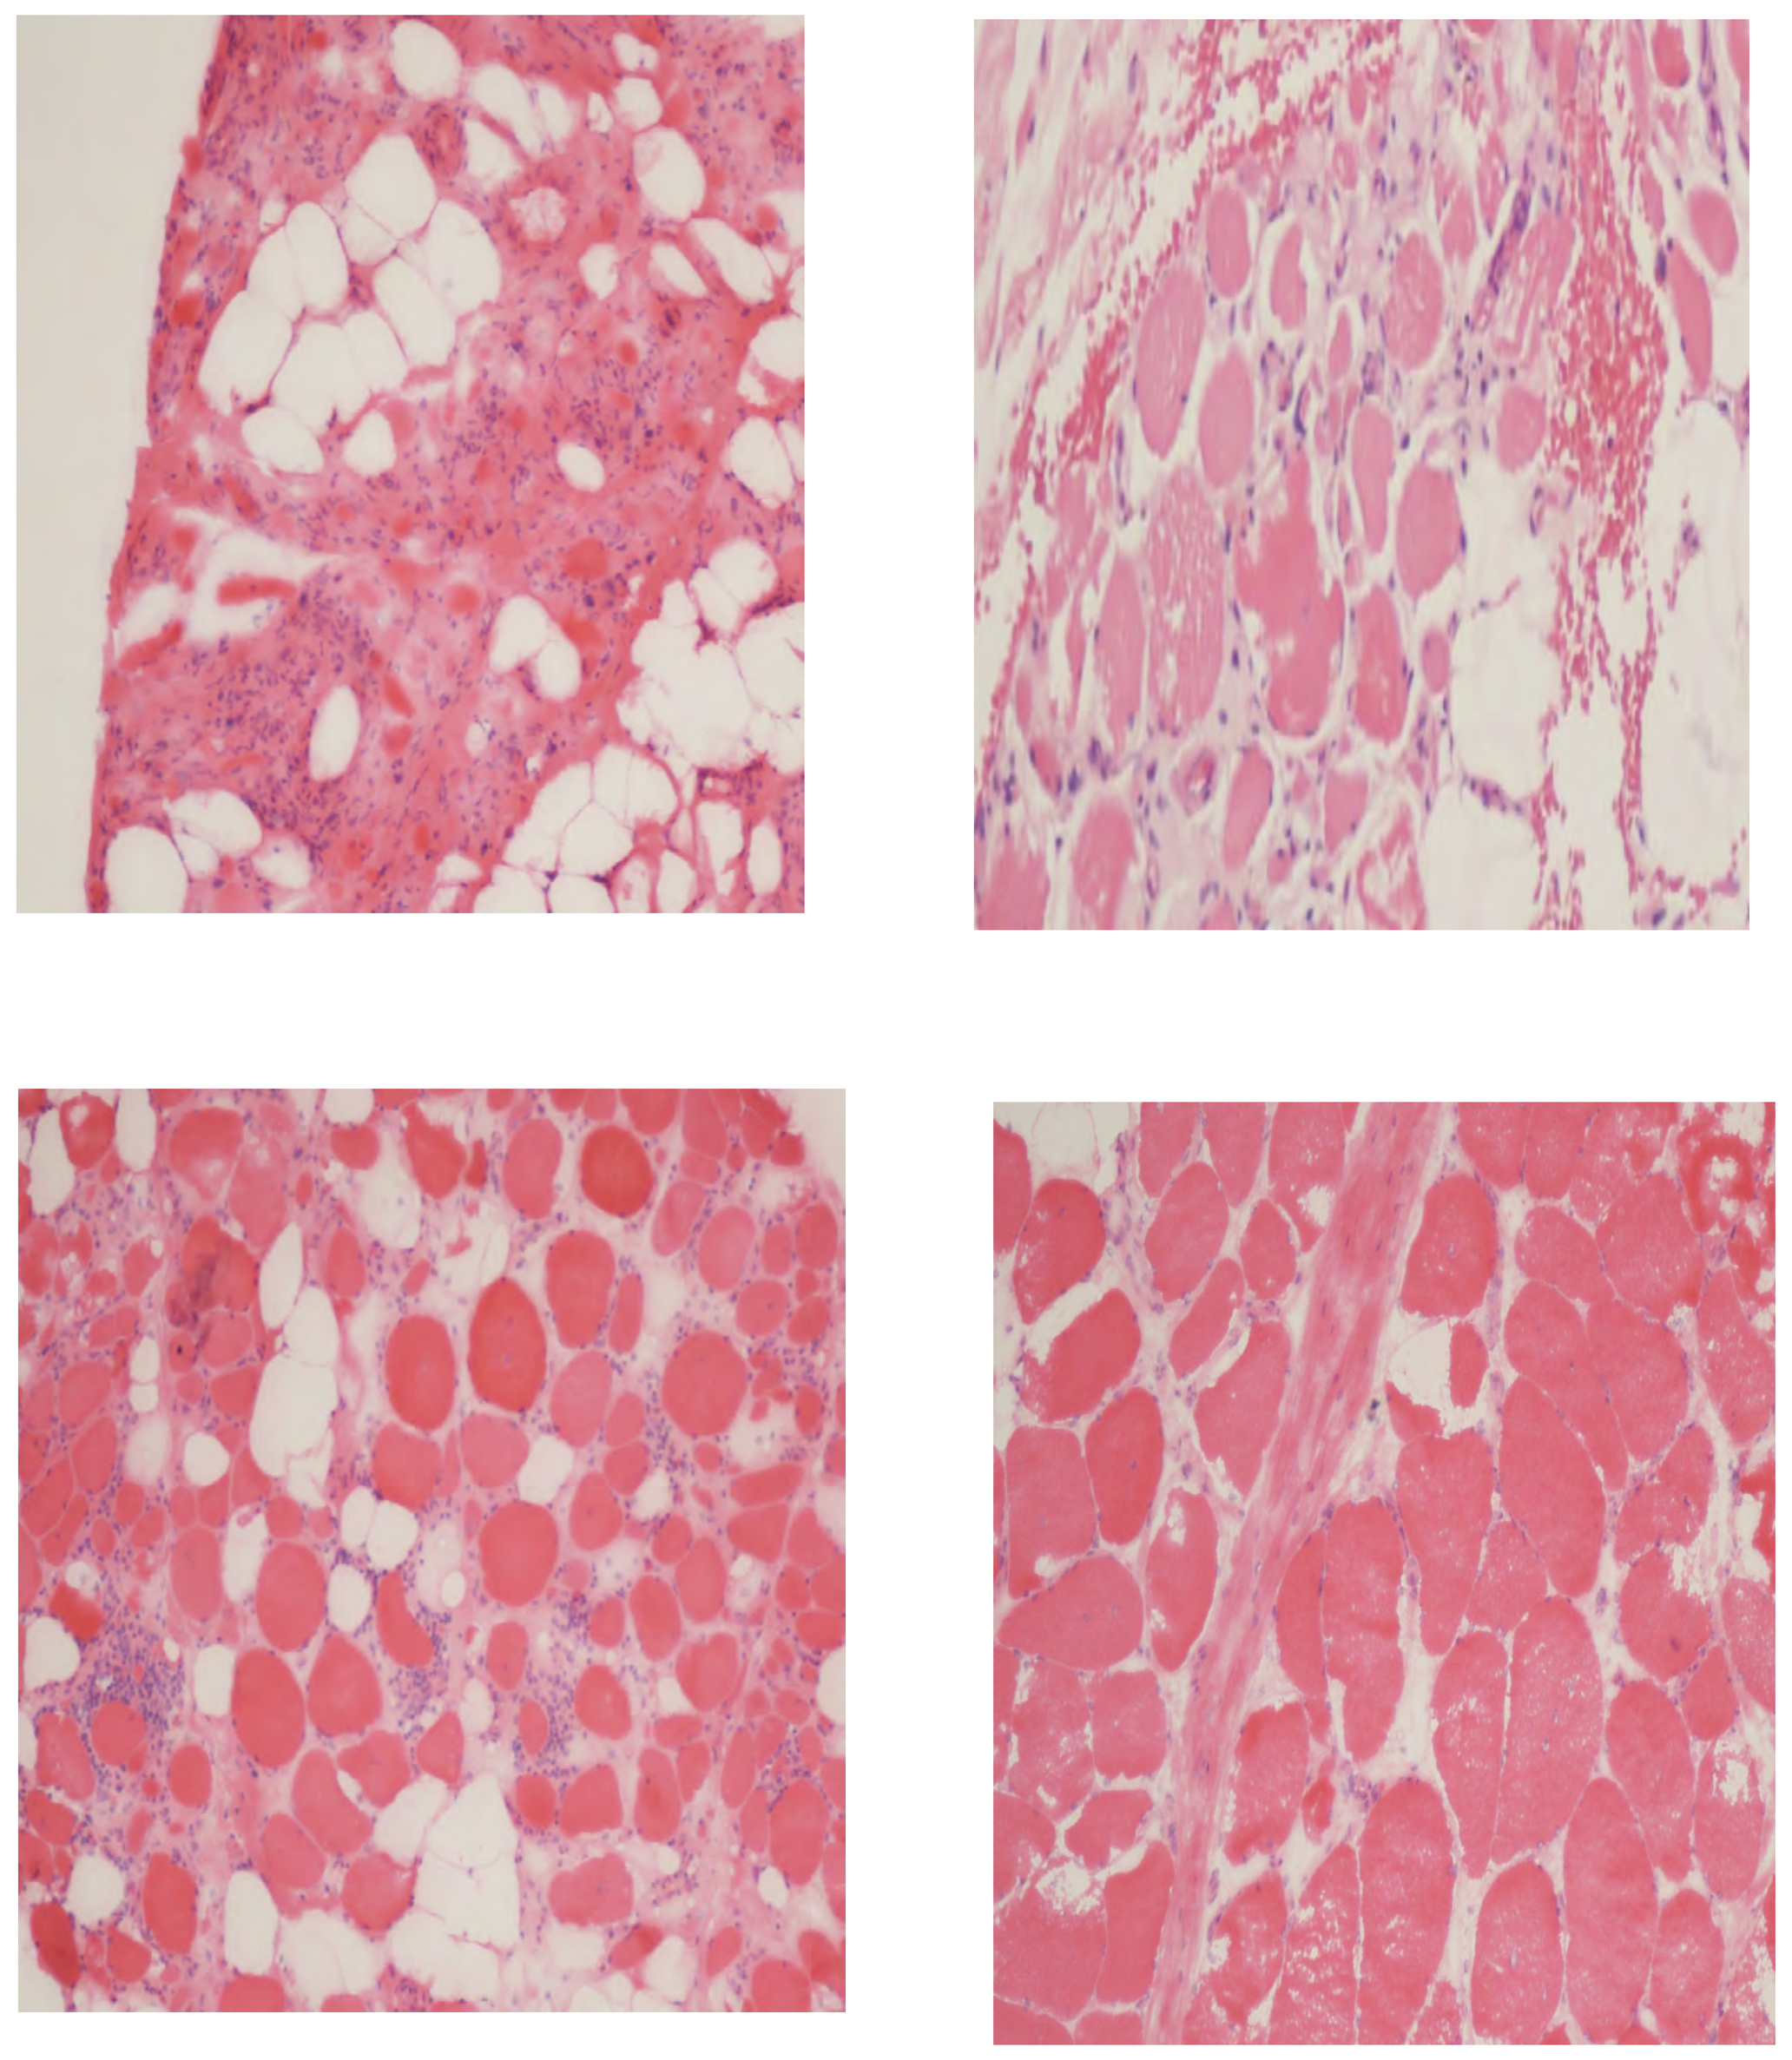

Figure 3. Biopsies on left are from the quadriceps before therapy with natalizumab and demonstrate significant endomysial inflammation. Biopsies on the right are the same patients after 6 months of therapy with marked reduction in endomysial inflammation

Conclusions

This poster describes the first two patients with IBM treated with Natalizumab. Natalizumab therapy was well tolerated for six months and there were no significant side effects.
Patients were moderately to severely affected at baseline and although both patients exhibited a mild improvement after three months of therapy by 6 months of therapy there was no significant change in muscle strength as measured by MMT or quantitative dynomometry.
Pre and post muscle biopsies however showed a marked reduction in endomysial inflammation. This reduction in inflammatory cells in the muscle biopsy validates the proposed mechanism of action of natalizumab. By binding to VLA-4 on the surface of the leukocytes, this monoclonal antibody prevents adherence to VCAM-1 found on the endothelial surface of the blood vessels and thereby prevents migration of inflammatory cells into the muscle tissue.
Our plans are to finish enrolling 4 more patients to complete this study, but the data from these two patients suggest a number of future hypotheses. If IBM is not a primarily autoimmune mediated disease than even if the inflammatory component of the disease is eliminated the disease may not improve. This was shown in a study by Dalakas using steroids and IVIG in patients with IBM. (1) One could also suggest that in a slowly progressive disease six months of therapy was not enough time to see a response.

Secondly, we suggest that the ability to turn off the inflammatory component may argue that a drug such as natalizumab may be more effective in autioimmune myopathies such as polymyositis or dermatomyositis than IBM. The potential benefit however needs to be weighed against the possible risk of immune suppression and the potential for the development of progressive multifocal leukoencephalopathy in patients who are JC virus positive (2).

This study was supported by Biogen.

1. Dalakas et al. Neurology 2001. 56: 323-327
2.Yousry TA, NEJM 2006:354, 924-933

printed by MegaPrint Inc. www.postersession.com